Comprehensive profiling and molecularly guided therapy (MGT) for carcinomas of unknown primary (CUP): CUPISCO – A Phase II, randomised, multicentre study comparing targeted therapy or immunotherapy with standard platinum-based chemotherapy

445TiP

Alwin Krämer,[1] Ferran Losa,[2] Laurie M. Gay,[3] Damian R. Page,[4] Stefan Foser,[4] Tariq I. Mughal,[3] Jeffrey S. Ross,[3] Giulia Baciarello,[5] Linda R. Mileshkin,[6] Stuart Osborne,[7] Holger Moch[8]

[1]Clinical Cooperation Unit, Molecular Hematology/Oncology, German Cancer Research Center (DKFZ) and Department of Internal Medicine V, University of Heidelberg, Heidelberg, Germany; [2]Medical Oncology Department, Hospital Sant Joan Despí – Moisès Broggi, Barcelona, Spain; [3]Pathology Group, Foundation Medicine, Inc., Cambridge, MA, USA; [4]Personalised Healthcare, F. Hoffmann-La Roche Ltd, Basel, Switzerland; [5]Department of Medical Oncology, Gustave Roussy, Villejuif, France; [6]Medical Oncology, Peter MacCallum Cancer Centre, Melbourne, VIC, Australia; [7]PDMA Operations (Biometrics), F. Hoffmann-La Roche Ltd, Basel, Switzerland; [8]Department of Pathology and Molecular Pathology, University Hospital Zurich, Zurich, Switzerland

BACKGROUND

- Carcinomas of unknown primary (CUP) – histologically confirmed metastatic cancers for which a standardised diagnostic work-up fails to identify the site of origin at the time of diagnosis (Pavlidis & Fizazi 2009; Fizazi et al., 2015) – are heterogeneous tumours of diverse origins and poor prognoses (Massard et al., 2011)
- Treatment of solid tumours is conventionally based on the tissue of origin; therefore, CUP ambiguity of origin makes treatment difficult
 - ESMO has developed a two-step algorithm for the treatment of CUP (Fizazi et al., 2015):
 1. If examination results, including clinical features, immunohistochemistry, radiology, laboratory values and additional diagnostic measures beyond those of the standard diagnostic work-up, strongly suggest a tissue of origin, treatment is initiated based on known site-specific therapies for the identified cancer type
 2. Patients for whom a likely tissue of origin cannot be posited are classified into two distinct clinicopathological subgroups (favourable-prognosis and **poor-prognosis CUP**). Subsequent treatment is initiated based on this classification (specific treatment for favourable-prognosis CUP and **two-drug platinum-based chemotherapy or chemotherapy/best supportive care for poor-prognosis CUP**)
- With the advent of large-scale DNA sequencing technologies, and the availability of a growing collection of targeted agents and immunotherapies, a new and rationally designed treatment paradigm may now be possible for CUP that is independent of tissue of origin and customised to the patient. However, prospective clinical studies evaluating this potentially promising approach are lacking

CUPISCO STUDY DESIGN

- The CUPISCO study (NCT03498521) is a Phase II, randomised, open-label, active-controlled, multicentre trial to directly assess whether molecularly guided therapy (MGT), based on comprehensive genomic profiling, is superior to recommended systemic chemotherapy in patients with **poor-prognosis CUP** who have achieved disease control after receiving 3 cycles of first-line platinum-doublet induction chemotherapy (Figure 1; eligibility criteria are shown in Table 1)
- Following induction therapy, patients are categorised as follows:
 - Category 1 patients achieved disease control (complete response, partial response or stable disease) after 3 cycles of first-line platinum-doublet induction chemotherapy
 - Category 2 patients experienced disease progression during 3 cycles of platinum-doublet induction chemotherapy
 - Category 1 patients will be randomised and Category 2 patients will go directly to targeted therapy (as they progressed on chemotherapy)
- All patients receive hybrid capture-based comprehensive genomic profiling (FoundationOne®, FoundationOne® Liquid; Foundation Medicine, Inc., Cambridge, MA, USA) to assess tumour genomic alterations, microsatellite instability and tumour mutational burden
 - FoundationOne® is an analytical, clinically validated single platform with molecular profiling of over 300 genes known to drive cancer growth. This genomic DNA assay has been shown to identify, with high selectivity and sensitivity, actionable mutations in CUP tumour specimens (Frampton et al., 2013)
 - FoundationOne® Liquid is a validated blood-based circulating tumour-DNA assay for solid tumours that interrogates 70 cancer-related genes (Clark et al., 2018)
- The primary endpoint of the CUPISCO study is progression-free survival (PFS) in patients who achieved disease control after receiving 3 cycles of platinum-doublet induction chemotherapy (Category 1 patients). The primary comparison is between MGT (pooled) and standard chemotherapy. This and other endpoints are shown in Table 2

Role and conduct of the Molecular Tumour Board

- A key element of the study design is a 'Molecular Tumour Board' (MTB), comprising the investigator, reference pathologist, reference oncologist and, when appropriate, a cancer genomics consultant, who advise on therapy choice for Category 1 patients randomised to MGT or for Category 2 patients based on tumour genomic profiles (Table 3)
- After evaluation at the central reference pathology laboratory, all submitted cases are classified as eligible or non-eligible for the CUPISCO study. This decision is communicated to the referring oncologist, the local pathologist and the local investigator
- Potentially actionable mutations in each patient's FoundationOne® report can be categorised into 'confirmed', 'experimental' and 'ineligible' for the purposes of therapy selection
- The FoundationOne® report will be shared with the referring oncologist and, if required, will be discussed during preparatory MTB meetings
- Relevant patient information will be reviewed simultaneously by all members of the MTB at a virtual MTB meeting, and a patient-specific treatment regimen will be decided upon by the treating investigator in consultation with the other board members
- During the above process, all treatment choices will be made according to the guidance provided in Table 3
- It is expected that choice of MGT may be ambiguous in some cases. The MTB will be provided with a charter containing guidance and rationales for therapy selection under such circumstances

Figure 1. CUPISCO study design

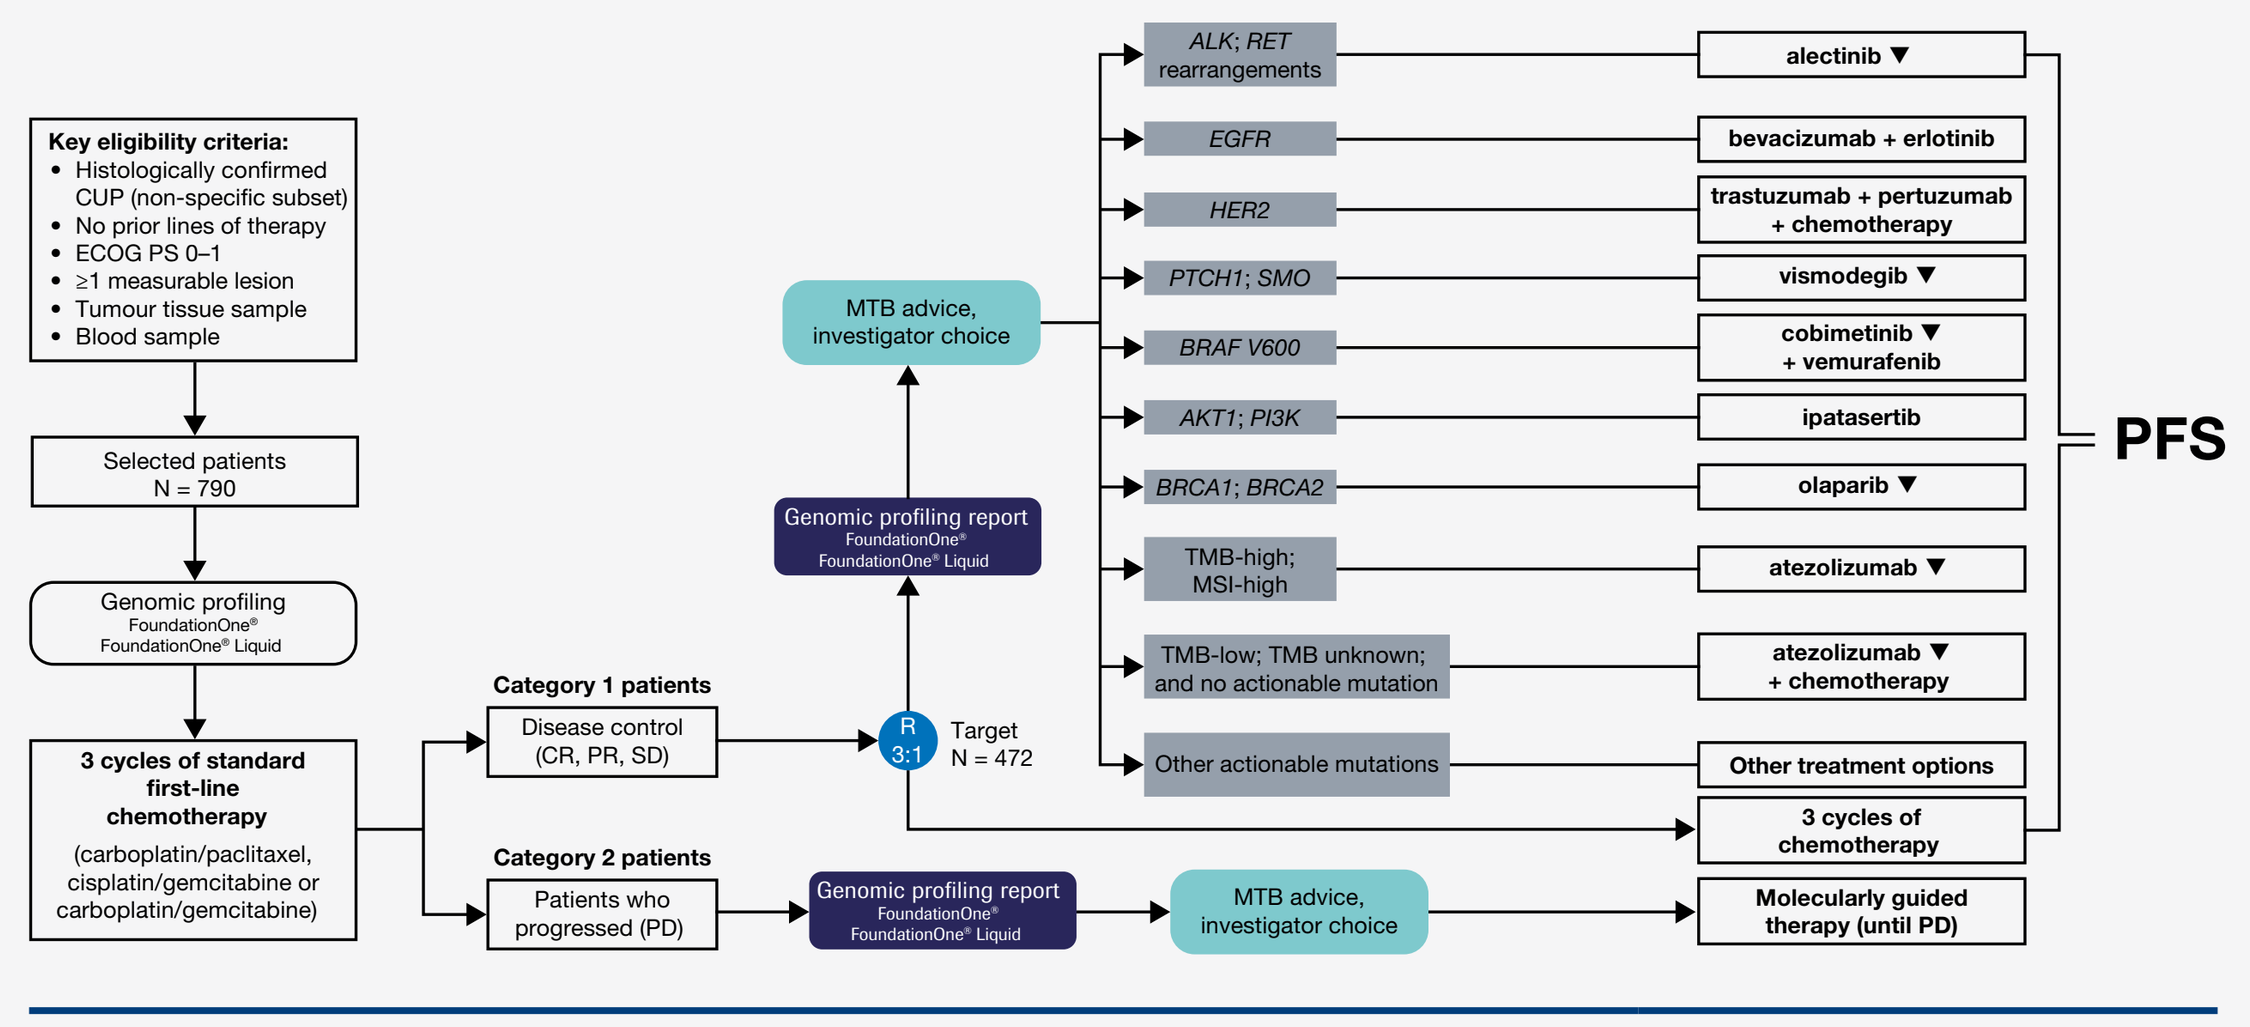
Key eligibility criteria:
• Histologically confirmed CUP (non-specific subset)
• No prior lines of therapy
• ECOG PS 0–1
• ≥1 measurable lesion
• Tumour tissue sample
• Blood sample
Selected patients N = 790
Genomic profiling FoundationOne® FoundationOne® Liquid
3 cycles of standard first-line chemotherapy (carboplatin/paclitaxel, cisplatin/gemcitabine or carboplatin/gemcitabine)
Category 1 patients
Disease control (CR, PR, SD)
Category 2 patients
Patients who progressed (PD)
R 3:1
Target N = 472
Genomic profiling report FoundationOne® FoundationOne® Liquid
MTB advice, investigator choice
ALK; RET rearrangements
EGFR
HER2
PTCH1; SMO
BRAF V600
AKT1; PI3K
BRCA1; BRCA2
TMB-high; MSI-high
TMB-low; TMB unknown; and no actionable mutation
Other actionable mutations
alectinib ▼
bevacizumab + erlotinib
trastuzumab + pertuzumab + chemotherapy
vismodegib ▼
cobimetinib ▼ + vemurafenib
ipatasertib
olaparib ▼
atezolizumab ▼
atezolizumab ▼ + chemotherapy
Other treatment options
3 cycles of chemotherapy
Molecularly guided therapy (until PD)
PFS

Randomisation is stratified by gender and response during the induction period (CR + PR vs SD). Genes listed comprise the confirmed selection of variants. Experimental genes may also be used for therapy selection. Therapies marked with ▼ are subject to additional monitoring. Reporting suspected adverse reactions after authorisation of the medicinal product is important. Adverse events should be reported to your respective local office: AstraZeneca for olaparib; Roche for alectinib, atezolizumab, cobimetinib and vismodegib. CUP, carcinoma of unknown primary; CR, complete response; ECOG PS, Eastern Cooperative Oncology Group performance status; MTB, Molecular Tumour Board; PD, progressive disease; PFS, progression-free survival; PR, partial response; R, randomisation; SD, stable disease; TMB, tumour mutational burden.

Table 1. Key eligibility criteria for the CUPISCO study

Key inclusion criteria
At study entry
Age ≥18 years at time of signing Informed Consent Form
Histologically and central laboratory-confirmed metastatic or advanced unresectable CUP diagnosed according to the criteria defined in the 2015 ESMO Clinical Practice Guidelines for CUP (Fizazi et al., 2015). Acceptable histologies: • Adenocarcinoma • Poorly differentiated adenocarcinoma • Poorly differentiated carcinoma The disease should not be amenable to resection and/or irradiation with curative intent during the course of the study
At least one lesion that is measurable according to RECIST v1.1
A tumour tissue sample that is suitable for: 1) the initial diagnosis of CUP at the study site's local laboratory, and 2) confirmation of the CUP diagnosis and generation of a FoundationOne® comprehensive genomic profile at a central reference pathology laboratory. If, after local diagnosis of CUP, insufficient tumour tissue remains for the central pathology laboratory to confirm the CUP diagnosis and generate a FoundationOne® profile, a fresh biopsy sample must then be collected during the screening period that meets the study's requirements
A blood sample suitable for analysis of circulating tumour DNA using the FoundationOne® Liquid assay
Eastern Cooperative Oncology Group performance status 0–1
No prior systemic therapy for the treatment of CUP
Life expectancy ≥12 weeks
Prior to start of therapy in the treatment period
Adequate haematological and end-organ function
Recovery from significant toxicity from platinum-doublet therapy to Grade ≤1, except for alopecia and for neurosensory toxicity, which must be Grade ≤2
Recovery from active infections requiring intravenous antibiotics, with antibiotic therapy ceased for ≥7 days prior to planned start of therapy

Key exclusion criteria
At study entry
Non-epithelial cancer; squamous-cell CUP
Patients belonging to any of the following subsets of CUP with favourable prognoses: • Poorly differentiated carcinoma with midline distribution • Women with papillary serous adenocarcinoma restricted to the peritoneal cavity • Women with adenocarcinoma involving only the axillary lymph nodes • Squamous-cell carcinoma of the cervical lymph nodes • Neuroendocrine tumours • Men with blastic bone metastases and elevated prostate-specific antigen • Patients with a single, small, potentially resectable tumour • CUP restricted to a single site • Colon cancer-type CUP
History of malignancy other than CUP within 5 years prior to screening, with the exception of malignancies with a negligible risk of metastasis or death (e.g., 5-year overall survival rate >90%), such as, but not limited to, adequately treated carcinoma *in situ* of the cervix, non-melanoma skin cancer, localised prostate cancer, ductal carcinoma *in situ*, or stage I uterine cancer
Treatment with investigational therapy within 28 days prior to initiation of study treatment
Ineligible for platinum-based chemotherapy
Any other disease, metabolic dysfunction, physical examination finding, or clinical laboratory finding that contraindicates the use of an investigational drug, may affect the interpretation of the results, or may render the patient at high risk of treatment complications
Prior to start of therapy in the treatment period
Known allergy or hypersensitivity to any component of the molecularly targeted agents
For all oral therapies: history of malabsorption syndrome, lack of physical integrity of the upper gastrointestinal tract, or other condition that would interfere with enteral absorption or result in the inability or unwillingness to swallow pills

CUP, carcinoma of unknown primary; RECIST v1.1, Response Evaluation Criteria In Solid Tumors, version 1.1.

Table 2. Objectives and endpoints in the CUPISCO study

Efficacy objective	Corresponding endpoints
To evaluate the efficacy of molecularly guided therapy versus platinum-based chemotherapy in patients with CUP whose best response to 3 cycles of platinum-doublet induction chemotherapy was confirmed CR, PR or SD	• Primary endpoint: — PFS1, defined in Category 1 patients as the time from randomisation to the first occurrence of disease progression, as assessed by the investigator according to RECIST v1.1, or death from any cause; whichever occurs first • Secondary endpoints: — OS, defined as the time from randomisation to death from any cause — ORR1, defined in Category 1 patients as the proportion of randomised patients who exhibit a CR or PR to molecularly guided therapy on two consecutive occasions ≥4 weeks apart — DCB1, defined in Category 1 patients as the time from the first occurrence of a CR, PR or SD after randomisation until disease progression or death from any cause; whichever occurs first — Responses will be determined by the investigator according to RECIST v1.1
Safety objective	**Corresponding endpoints**
To evaluate the safety of molecularly guided therapy in all patients with CUP who receive targeted therapy or cancer immunotherapy	• Incidence, nature and severity of adverse events • Incidence and reasons for any dose reductions, interruptions, or premature discontinuation of any component of study treatment • Clinically significant laboratory values and vital signs
Exploratory objectives	**Corresponding endpoints**
To evaluate the efficacy of molecularly guided therapy in patients with CUP who progressed during 3 cycles of platinum-doublet induction chemotherapy	PFS2, OS2, ORR2 and DCB2, assessed by the investigator in Category 2 patients according to RECIST v1.1, as described above
To evaluate the mutagenic effects of 3 cycles of platinum-doublet induction chemotherapy in all patients with CUP	Genomic profiles pre- and post-platinum-doublet induction chemotherapy, as assessed using FoundationOne® Liquid
To evaluate clonal evolution of CUP during targeted or cancer immunotherapy treatment	Genomic profiles pre-treatment and at disease progression in patients receiving targeted therapy or cancer immunotherapy
To characterise CUP tumours and their microenvironment on a molecular level	Molecular profiling by, e.g., gene expression and immunohistochemistry analyses
To evaluate the HRQoL effects of molecularly guided therapy in patients with CUP whose best response to 3 cycles of platinum-doublet induction chemotherapy was confirmed CR, PR or SD	Absolute change from randomisation in FACT-G score in Category 1 patients Absolute change from randomisation in HADS score in Category 1 patients
To evaluate health status utility of molecularly guided therapy in patients with CUP whose best response to 3 cycles of platinum-doublet induction chemotherapy was confirmed CR, PR or SD	Absolute change from randomisation in EQ-5D-5L index-based score in Category 1 patients

CR, complete response; CUP, cancer of unknown primary; DCB, duration of clinical benefit; EQ-5D-5L, EuroQol 5-Dimensional 5-Level questionnaire; FACT-G, Functional Assessment of Cancer Therapy: General; HADS, Hospital Anxiety and Depression Scale; HRQoL, health-related quality of life; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PR, partial response; RECIST v1.1, Response Evaluation Criteria in Solid Tumours, version 1.1; SD, stable disease.

Table 3. Identified genomic alterations and corresponding treatment options in the CUPISCO study

Molecularly guided therapies	Identified confirmed actionable alterations
Targeted therapies	
Alectinib	*ALK*, *RET* rearrangements
Vismodegib	Inactivating *PTCH1*, activating *SMO* alterations
Ipatasertib	*AKT1*, *PI3K* actionable alterations
Olaparib	*BRCA1*, *BRCA2* or homologous recombination deficiency based on loss of heterozygosity
Erlotinib + bevacizumab	*EGFR* actionable alterations
Vemurafenib + cobimetinib	*BRAF V600* alterations
Subcutaneous trastuzumab + pertuzumab + chemotherapy[1]	*HER2* actionable alterations
Immunotherapy	
Atezolizumab	TMB-high (≥16 mutations/Mb), MSI-high (Chalmers et al., 2017)
Atezolizumab + chemotherapy[1]	TMB-low or unknown (<16 mutations/Mb) in patients without actionable mutations
Other treatment options	**Potential rationales**
Alternative therapies (only if the investigator in consultation with the MTB has strong evidence to support a therapy not represented in the nine investigational treatment arms above)	• Strong suspicion of a primary tumour revealed by comprehensive genomic profiling • Strong rationale for alternative, commercially available, targeted therapy • Negative predictor of response to anti–PD-1 or anti–PD-L1 agents • Patients who do not have a genetic alteration allowing for assignment to a protocol-mandated targeted therapy and who are contraindicated for atezolizumab

[1] Continuation of the initial platinum-doublet induction chemotherapy.
MTB, Molecular Tumour Board; PD-1, programmed death-1; PD-L1, programmed death-ligand 1; TMB, tumour mutational burden.

KEY ASSESSMENTS

- Response will be assessed by the investigator on the basis of physical examinations, computerised tomography scans and magnetic resonance imaging, using Response Evaluation Criteria In Solid Tumors (version 1.1) at the end of the induction period, every 3 treatment cycles, and every 3 months during follow-up. An objective response should be confirmed by repeat assessments ≥4 weeks after initial documentation
- Adverse events (AEs) will be monitored and documented continuously during the study, and serious AEs will also be documented and reported, as will AEs of special interest. All AEs will be graded according to the National Cancer Institute's Common Terminology Criteria for Adverse Events (version 4.0)

KEY STATISTICAL ANALYSES

- The primary analysis of PFS will be based on the intention-to-treat population (all randomised patients, whether or not the assigned study treatment was received)
 - Stratified analysis of the primary endpoint will be completed according to the predefined randomisation stratification factors (gender and response to platinum-doublet induction chemotherapy)
 - Kaplan–Meier methodology will be used to estimate the median PFS for MGT (pooled) and standard chemotherapy, together with the 95% confidence interval
 - PFS will be compared between MGT (pooled) and standard chemotherapy by the stratified log-rank test
 - The hazard ratio for PFS will be estimated using a Cox proportional hazards model
- At the time of the primary analysis of PFS, an interim analysis of overall survival will be performed
- An estimate of objective response rate will be calculated for MGT (pooled) and standard chemotherapy, and its 95% confidence interval will be calculated using the Clopper–Pearson method
 - Objective response rate will be compared between MGT (pooled) and standard chemotherapy using the stratified Cochran–Mantel–Haenszel test
 - The stratification factors will be the same as those for the analysis of the primary endpoint
- Safety analyses will be descriptive

ENROLMENT

- Enrolment of 790 patients is planned across 23 countries (Figure 2) and ~101 sites
- Recruitment is ongoing; four patients have been enrolled to date and three are currently in screening

Figure 2. Countries enrolling patients in the CUPISCO study

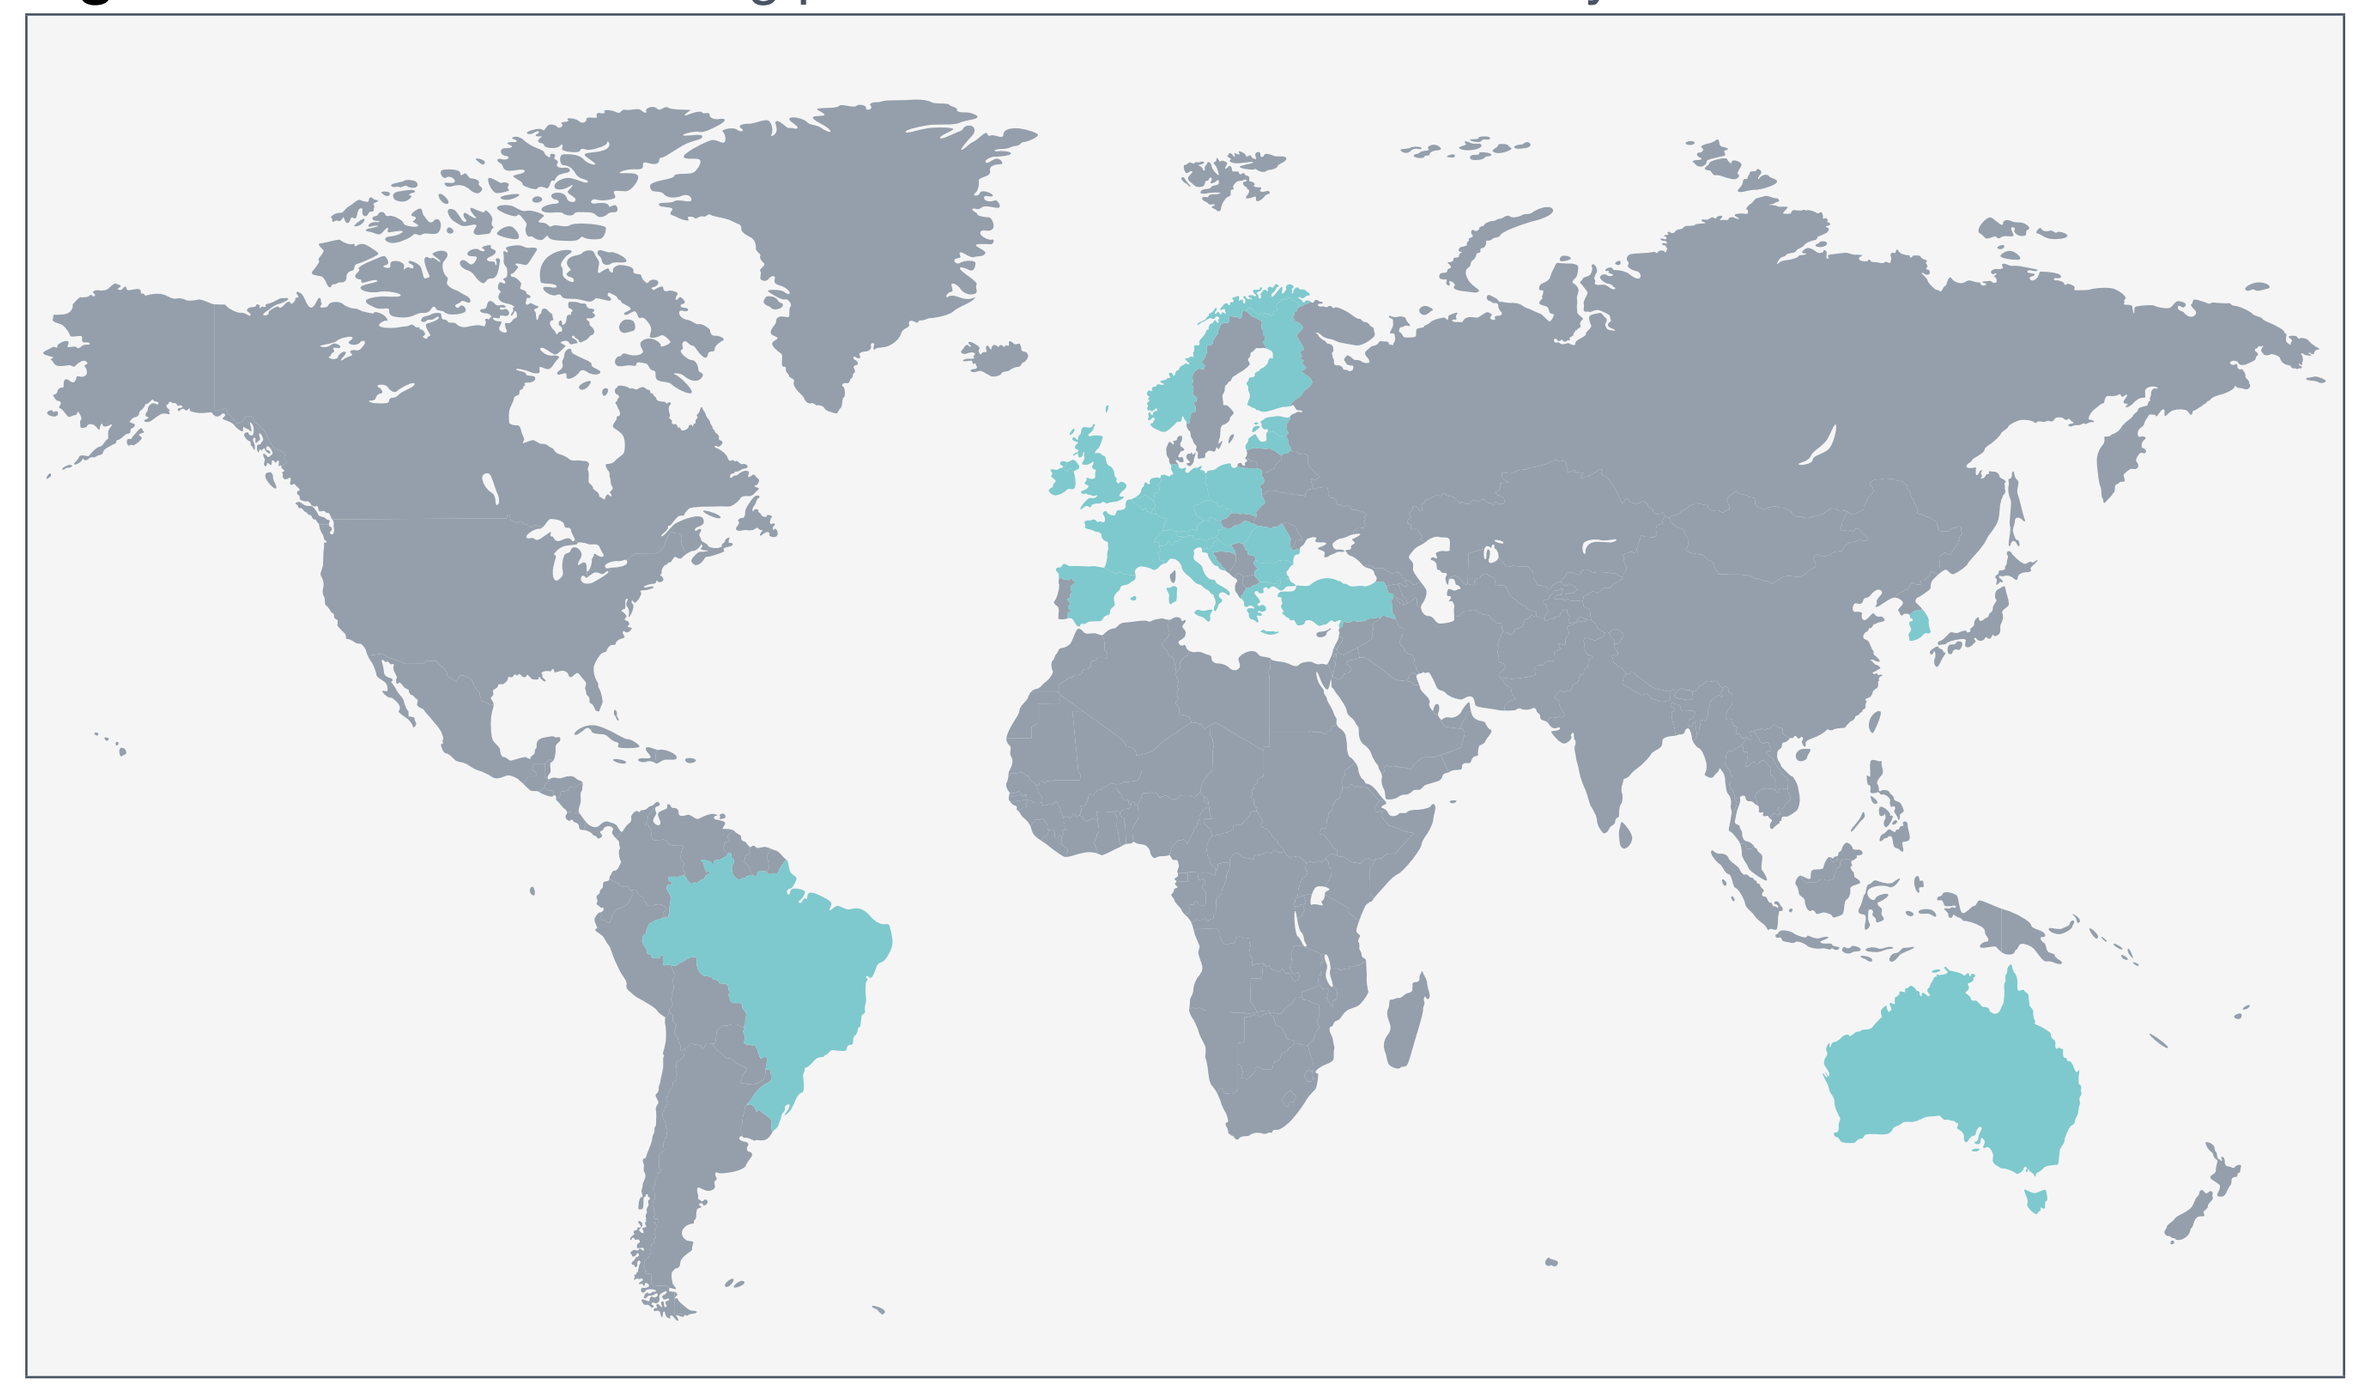

CONTACT INFORMATION FOR THE CUPISCO STUDY

- Email: katarzyna.stadnicka@roche.com
- Phone: +41 61 68 74 494
- ClinicalTrials.gov: https://clinicaltrials.gov/ct2/show/NCT03498521

REFERENCES

Chalmers ZR, et al. Genome Med 2017;9:34.
Clark TA, et al. J Mol Diagn 2018;20:686–702.
Fizazi K, et al. Ann Oncol 2015;26(Suppl 5):v133–v138.
Frampton GM, et al. Nat Biotechnol 2013;11:1023–1031.
Massard C, et al. Nat Rev Clin Oncol 2011;8:701–710.
Pavlidis N & Fizazi K. Crit Rev Oncol Hematol 2009;69:271–278.

ACKNOWLEDGEMENTS

Study sponsored by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

We would like to acknowledge all investigators, the MTB, and patients participating in this trial, as well as the CUPISCO study team, and Andreas Beringer of F. Hoffmann-La Roche Ltd for his contributions to this poster.

DRP's current affiliation: Roche Pharma (Schweiz) AG, Reinach, Switzerland.

TIM's current affiliation: Hematology and Oncology, Tufts University Medical Center, Boston, MA, USA.

Support for third-party writing assistance for this poster, furnished by Daniel Clyde, PhD, of Health Interactions, was provided by F. Hoffmann-La Roche Ltd.

CONFLICT OF INTEREST DECLARATION

Conflict of interest declaration: AK: Honoraria received from Roche and Bayer; consulting or advisory role for Roche, Bayer and Daiichi; research funding from Bayer; patents, royalties, other intellectual property from Bayer; travel, accommodation and expenses from Roche. FL: Meeting attendance for Sanofi, Merck, Amgen and Lilly; consultancy or lecture fees from Ferrer and Roche; scientific advisory board meeting attendance for Merck, Amgen, Sanofi and Servier; steering committee meeting attendance for FMI/Roche (for the CUPISCO study). LMG: Employment at Foundation Medicine, Inc.; ownership of stock or other interests at Gilead Sciences and Foundation Medicine, Inc. DRP: Employment at Roche/Genentech and immediate family member employment at Novartis; ownership of stock or other interests at Roche and immediate family member ownership of stock or other interests at Novartis. SF: Employment at Roche/Genentech; ownership of stock or other interests at Roche. TIM: Employment at Foundation Medicine, Inc.; leadership at Foundation Medicine, Inc.; stock or other ownership at Foundation Medicine, Inc.; honoraria received from Sanofi; patents, royalties, other intellectual property for Oxford University Press and Informa. JSR: Employment at Foundation Medicine, Inc.; leadership at Foundation Medicine, Inc.; ownership of stock or other interests at Foundation Medicine, Inc. GB: Consulting or advisory role for Janssen and Sanofi; travel, accommodation and expenses for Janssen, Astellas, AstraZeneca and Sanofi. LRM: Travel, accommodation and expenses provided by Roche and BeiGene. SO: Employed by Roche. HM: Consulting or advisory role for Roche and Definiens; research funding from Roche.

NB: There may be associated costs for downloading data. These costs may vary depending on your service provider and may be high if you are using your smartphone abroad. Please check your phone tariff or contact your service provider for more details. Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

FoundationOne® and FoundationOne® Liquid are registered trademarks of Foundation Medicine, Inc.
Roche is the licensed distributor of Foundation Medicine products outside of the United States.

Please contact the lead author at a.kraemer@Dkfz-Heidelberg.de for permission to reprint and/or distribute.
Presented at the European Society for Medical Oncology (ESMO) 2018 Congress, 19–23 October 2018, Munich, Germany.